This is a repository copy of *Cost-effectiveness analysis of nilotinib versus imatinib for the treatment of chronic phase philadelphia chromosome positive chronic myeloid leukaemia*.

White Rose Research Online URL for this paper:
https://eprints.whiterose.ac.uk/43907/

Conference or Workshop Item:

Mildred, M., Ward, S., Squires, H. et al. (1 more author) (2012) Cost-effectiveness analysis of nilotinib versus imatinib for the treatment of chronic phase philadelphia chromosome positive chronic myeloid leukaemia. In: 17th Congress of EHA, 14-17 Jun 2012, Amsterdam, The Netherlands.

Reuse

Items deposited in White Rose Research Online are protected by copyright, with all rights reserved unless indicated otherwise. They may be downloaded and/or printed for private study, or other acts as permitted by national copyright laws. The publisher or other rights holders may allow further reproduction and re-use of the full text version. This is indicated by the licence information on the White Rose Research Online record for the item.

Takedown

If you consider content in White Rose Research Online to be in breach of UK law, please notify us by emailing eprints@whiterose.ac.uk including the URL of the record and the reason for the withdrawal request.

eprints@whiterose.ac.uk
https://eprints.whiterose.ac.uk/

No: 104

Cost-Effectiveness Analysis of Nilotinib Versus Imatinib For The Treatment Of Chronic Phase Philadelphia Chromosome Positive Chronic Myeloid Leukaemia

Matthew Mildred MSc,[1,2] Sue Ward BA,[2] Hazel Squires MSc,[2] Elizabeth Gray MSc[3]

[1]Abacus International, Bicester, UK; [2]School of Health and Related Research (ScHARR), University of Sheffield, Sheffield, UK; [3]Novartis Pharmaceuticals UK Ltd, Frimley, Surrey, UK

BACKGROUND

- Nilotinib is a tyrosine kinase inhibitor (TKI) for the treatment of adult patients with newly diagnosed Philadelphia chromosome positive (Ph+) chronic myeloid leukaemia (CML) in chronic phase.
- The ENESTnd phase III trial demonstrated that nilotinib has clinical superiority over current standard treatment of first-line imatinib in patients with chronic phase Ph+ CML, on the basis that fewer patients progressed to accelerated phase/blast crisis.[1,2]
- Within this trial, significantly fewer patients progressed on nilotinib 300 mg BD (0.7% patients, p=0.003) or nilotinib 400 mg BD (1.8% patients, p=0.0089) compared to imatinib 400 mg OD (6.0% patients).[1,2]
- Whilst the clinical benefits of nilotinib have been demonstrated, the cost-effectiveness of first-line nilotinib has not been explored.

OBJECTIVES

- To evaluate the cost-effectiveness of first-line nilotinib compared to first-line imatinib for patients newly diagnosed with chronic phase Ph+ CML.
- **Population:** Adult patients with Ph+ CML diagnosed in chronic phase and who do not initially receive a stem cell transplant (SCT).
- **Intervention:** First-line nilotinib 300 mg BD, second-line dasatinib 100 mg OD.
- **Comparator:** First-line imatinib 400 mg OD, second-line dasatinib 100mg OD.
- **Outcomes:** Costs, life-years (LYs), quality-adjusted life-years (QALYs), incremental cost effectiveness ratios (ICERs).

METHODS

- A Markov state-transition model based on the 24 month follow-up from the ENESTnd randomised controlled trial was developed to simulate the transitions of a hypothetical cohort of chronic phase CML patients over a lifetime (Figure 1). The clinical effectiveness of first-line treatment was modelled by extrapolating time on treatment data from the ENESTnd trial using a Weibull curve.
- **Health-related quality of life:** EQ-5D utilities were applied to patients in each health state (Figure 1) and utility decrements were estimated for patients experiencing severe (grade 3 and 4) adverse events on TKI therapy (Table 1).
- **Costs:** Costs (Sterling, 2011) associated with the different drug therapies, allogeneic stem cell transplantation (allo-SCT), routine hospital appointments for administration and monitoring, and treatment for severe adverse events were included. A patient access scheme is available for first-line nilotinib therapy and was included in the analysis.
- **Perspective:** UK National Health Service (NHS) and Personal Social Services (PSS).
- **Discount rate:** Future costs and benefits were discounted by 3.5% to reflect society's time preference.
- **Probabilistic sensitivity analysis (PSA):** All uncertain model parameters were simultaneously sampled from their probability distribution in order to provide a mean estimate of the costs, survival and QALYs. Extensive one-way sensitivity analyses were also performed to evaluate the impact of varying baseline estimates and assumptions.

Table 1. Treatment costs, utilities and disutilities included in the model

Parameter	Value	Source
Costs		
Nilotinib 300 mg BD (28 days)	£2,432.85	[3]
Imatinib 400 mg OD (30 days)	£1,724.39	[4]
Dasatinib 100 mg OD (30 days)	£2,504.96	[4]
HU 500mg (25 days)	£10.47	[4]
Allogeneic SCT	£99,224.38	[5,6]
Utilities		
Chronic phase utility	0.854	[7]
Accelerated phase utility	0.595	[7]
Blastic phase utility	0.595	[7]
Transplant utility	0.813	Calculated*
Disutilities		
Nilotinib disutility	0.010	Calculated*
Imatinib disutility	0.016	Calculated*
HU disutility	0	Assumption
Dasatinib disutility	0.019	Calculated*

*Based on the frequency and duration of serious adverse events.
HU = hydroxyurea; SCT = stem cell transplantation.

Presented at the 17th Congress of EHA, Amsterdam, The Netherlands, 16th June 2012.

Figure 1. Treatment pathways and health states in cost-effectiveness model

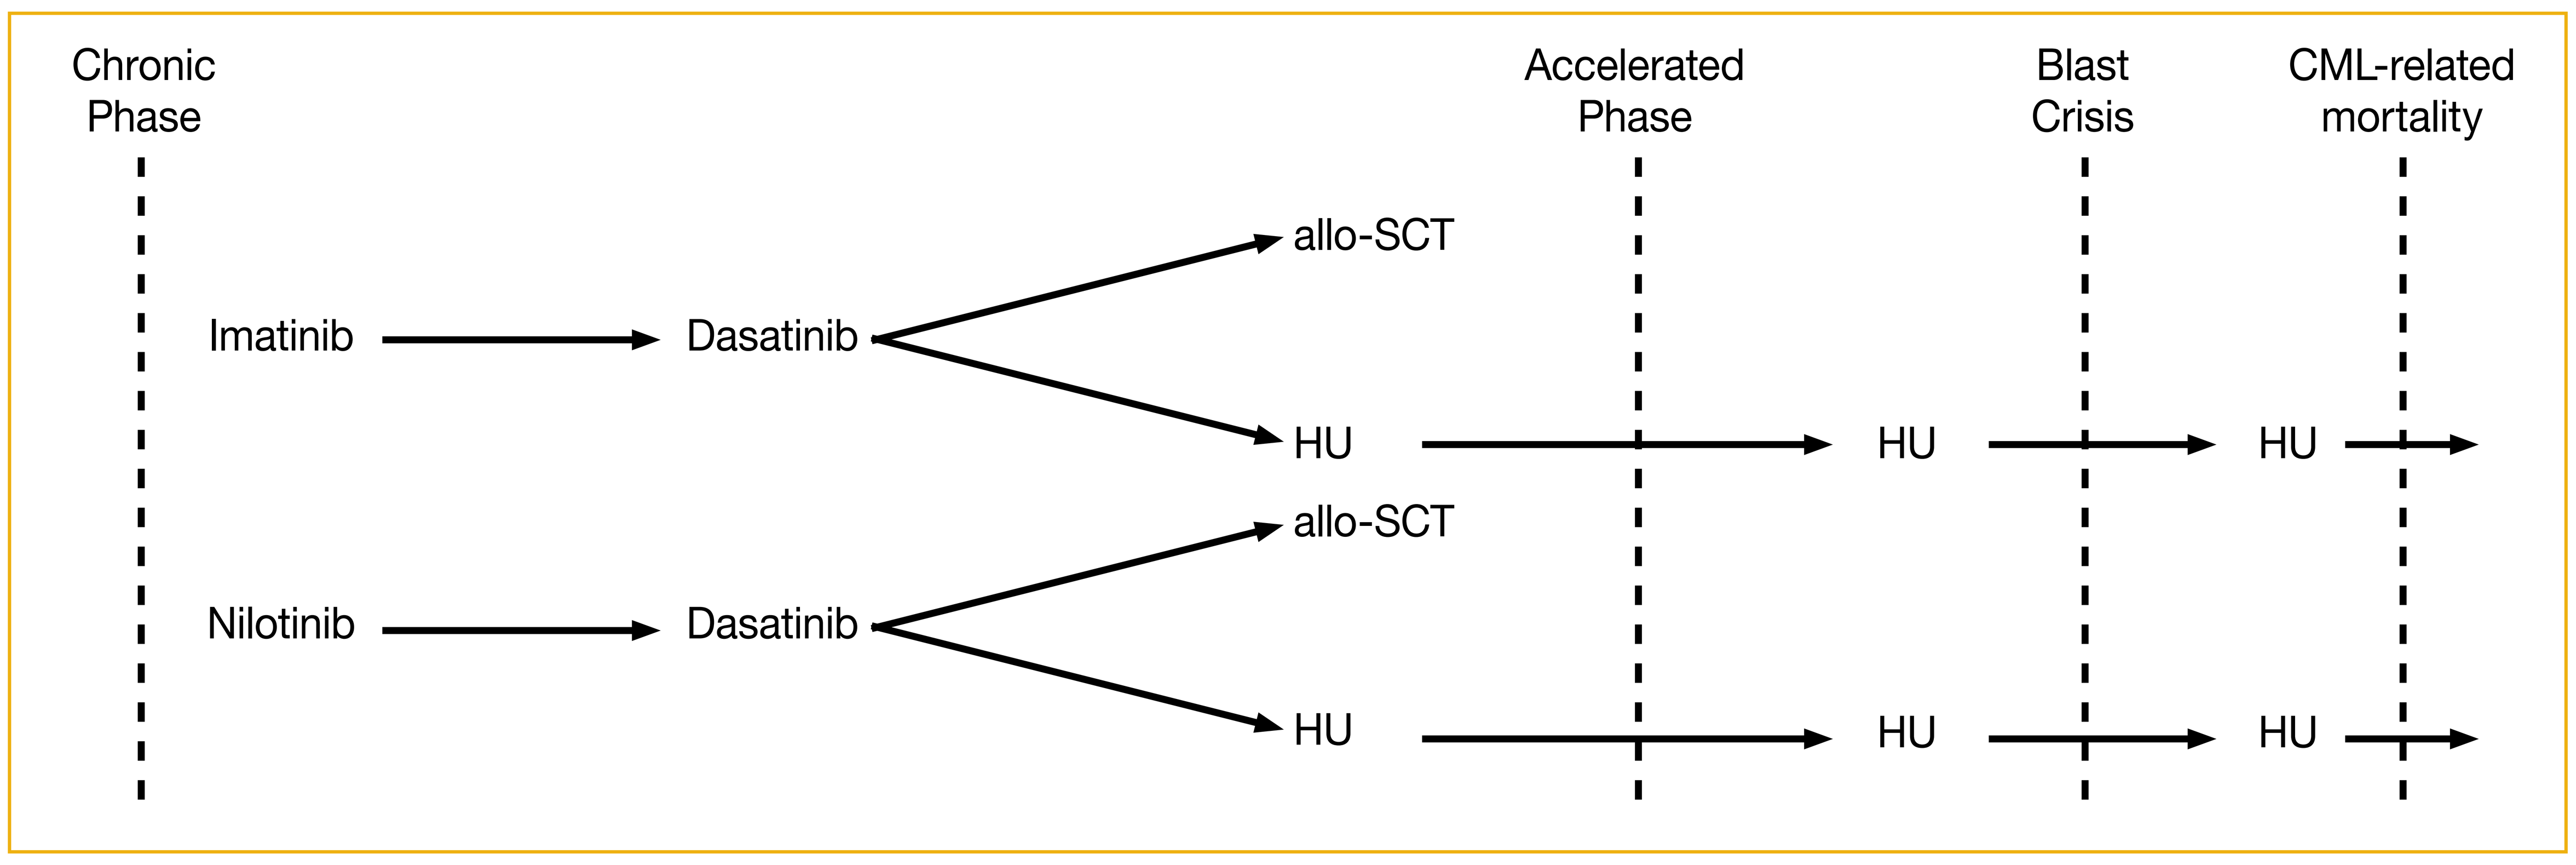
Chronic Phase
Accelerated Phase
Blast Crisis
CML-related mortality
Imatinib
Dasatinib
allo-SCT
HU
HU
HU
Nilotinib
Dasatinib
allo-SCT
HU
HU
HU

Patients may die from other causes at any time. CP = chronic phase; AP = accelerated phase; BC = blast crisis; allo-SCT = allogeneic stem cell transplantation; HU = hydroxyurea.

RESULTS

- Overall survival is estimated to be consistently greater in the nilotinib arm than the imatinib arm for all time points.
- Figure 2 presents the modelled overall survival of patients in the nilotinib arm and depicts the transition of patients through each of the health states. The orange area represents the number of patients alive on first-line nilotinib, with the blue dashed portion of the orange area representing the difference in numbers alive on first-line nilotinib compared to first-line imatinib.
- It can be seen that nilotinib has a slower rate of progression to worse disease health states. The overall effect is that nilotinib extends life in comparison to imatinib.

Figure 2. Estimated survival for patients receiving first-line nilotinib followed by second-line dasatinib

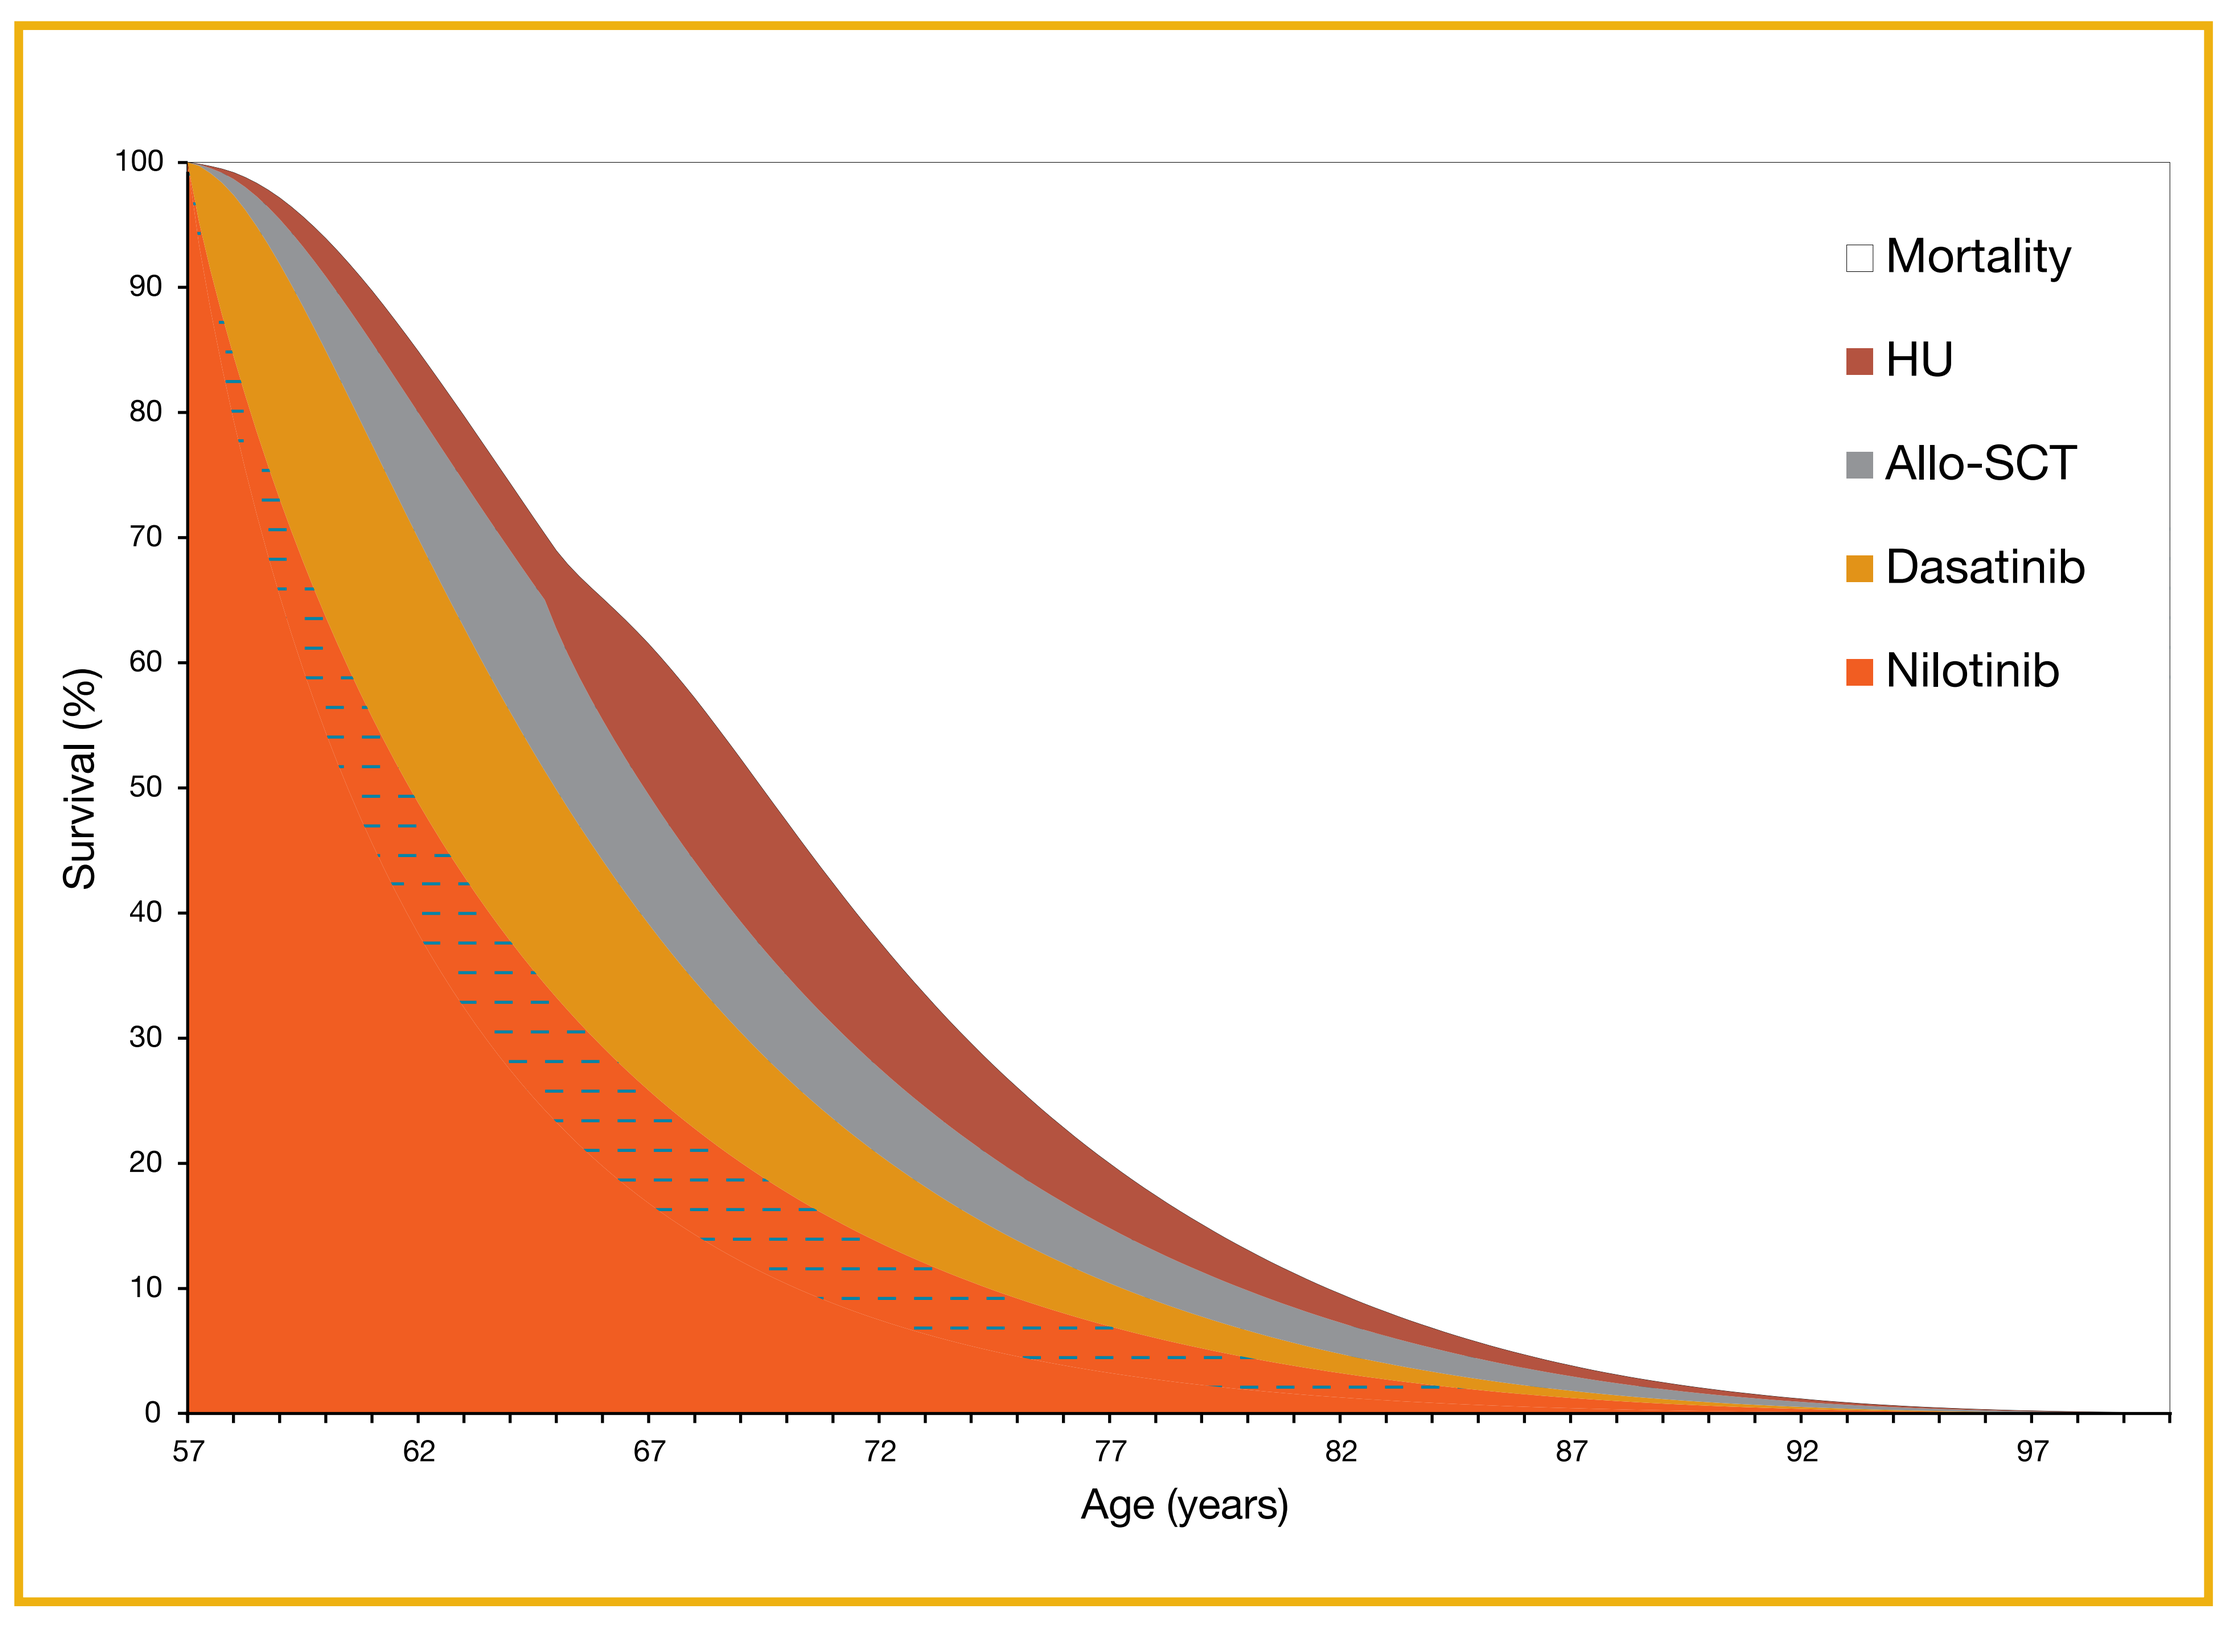
100
90
80
70
60
50
40
30
20
10
0
Survival (%)
57
62
67
72
77
82
87
92
97
Age (years)
Mortality
HU
Allo-SCT
Dasatinib
Nilotinib

- Patients receiving first-line nilotinib followed by second-line dasatinib are estimated to live an additional 0.64 years (33 weeks) compared to the imatinib arm, with an associated cost saving of £10,700 over a lifetime (Table 2).
- The mean undiscounted survival in the nilotinib arm is estimated to be 13.96 years compared to 13.32 years in the imatinib arm.
- After adjusting for quality of life, patients are estimated to gain an additional 0.49 QALYs in the nilotinib arm compared to the imatinib arm.
- After discounting, patients are estimated to accrue an additional 0.35 LYs and 0.28 QALYs in the nilotinib arm compared to the imatinib arm. Expected lifetime (discounted) costs in the nilotinib arm are £220,416 compared to £232,941 in the imatinib arm.

Table 2. Cost-effectiveness results

Parameter	Life-years	QALYs	Lifetime costs
Undiscounted			
Nilotinib	13.96	10.71	£279,000
Imatinib	13.32	10.22	£289,700
Difference	0.64	0.49	-£10,700
ICER	Dominated	Dominated	
Discounted			
Nilotinib	10.52	8.18	£220,500
Imatinib	10.17	7.90	£233,000
Difference	0.35	0.28	-£12,500
ICER	Dominated	Dominated	

Costs rounded to nearest hundred. ICER = incremental cost-effectiveness ratio; QALYs = quality-adjusted life-years.

- The nilotinib arm therefore dominates the imatinib arm as it is more effective and less costly.
- In all of the sensitivity analyses, first-line nilotinib remained cost-effective at a threshold of £20,000 per QALY gained compared with imatinib, with nilotinib dominating imatinib in the majority of cases.

CONCLUSIONS

- **Our analysis suggests that first-line nilotinib provides a cost-effective use of NHS resources for the treatment of chronic phase Ph+ CML.**
- **This is in line with recent guidance from the Scottish Medicines Consortium (SMC) and the National Institute for Health and Clinical Excellence (NICE), both of whom have recommended nilotinib as an option for the first-line treatment of adults with chronic phase Ph+ CML in Scotland and England respectively.[8,9]**

References

1. Kantarjian, H.M. et al. 2011. Nilotinib versus imatinib for the treatment of patients with newly diagnosed chronic phase, Philadelphia chromosome-positive, chronic myeloid leukaemia: 24-month minimum follow-up of the phase 3 randomised ENESTnd trial. The Lancet Oncology. 12 (9) pp.841-851.
2. Hochhaus, A. et al. 2011. Superior efficacy of nilotinib compared with imatinib in newly-diagnosed patients with chronic myeloid leukemia in chronic (CML-CP): ENESTnd minimum 24-month follow-up. Haematologica 96 (supplement 2), pp.203-204.
3. Monthly Index of Medical Specialities (MIMS). London: Haymarket Medical Media; March 2011.
4. British Medical Association. British National Formulary (BNF), No. 61. London: British Medical Association and the Royal Pharmaceutical Society of Great Britain; March 2011
5. Millen M, Phennah D. 2009. London Specialist Commissioning Group. Adult BMT costing and price model workshop. Held in London, 6 July 2009. 2009.
6. Ashfaq, K. et al. 2010. Clinical effectiveness and cost-effectiveness of stem cell transplantation in the management of acute leukaemia: a systematic review. Health Technol Assess, 14 pp.1-141.
7. Reed, S.D. et al. 2004. Cost-effectiveness of imatinib versus interferon- plus low-dose cytarabine for patients with newly diagnosed chronic-phase chronic myeloid leukemia. Cancer, 101 pp.2574-83.
8. Scottish Medicines Consortium (SMC). 2011. nilotinib (Tasigna), http://www.scottishmedicines.org.uk/SMC_Advice/Advice/709_11_nilotinib_Tasigna/nilotinib_Tasigna.
9. National Institute for Health and Clinical Excellence (NICE). 2012. Leukaemia (chronic myeloid, first line) - dasatinib, nilotinib and standard-dose imatinib (TA251), http://guidance.nice.org.uk/TA251.

Available online at http://eprints.whiterose.ac.uk/43907/
or scan with your smart phone:

This study was supported by Novartis Pharmaceuticals UK Ltd.